ASCO 2026

2605

Session:

Developmental Therapeutics - Immunotherapy

BI-1808 + Pembrolizumab: Responses to a Chemotherapy-free Regimen in Advanced Ovarian Cancer

Rebecca Kristelleit[A], Anja Williams[B], Juanita Lopez[C], Jeffrey Yachnin[D], Ana Carneiro[E], Kristoffer Rohrberg[F], Rikke Løvendahl Eefsen[G], Harriet Walter[H], Sean Lim[I], Marica Vaapil[J], Susanne Gertsson[J], Petra Holmkvist[J], Ingrid Karlsson[J], Danijela Lindahl[J], Linda Mårtensson[J], Dmytro Piliuhin[J], Anette Sundstedt[J], Ingrid Teige[J], Johan Wallin[J], Michael Chisamore[K], Björn Frendeus[J,L], Andres McAllister[J]

[A] Guy's and St Thomas' NHS Foundation Trust, United Kingdom, [B] Sarah Cannon Research Institute, United Kingdom, [C] The Royal Marsden NHS Foundation Trust, Institute of Cancer Research, United Kingdom, [D] Karolinska Comprehensive Cancer Center, Karolinska Universitetssjukhuset, Solna, Sweden, [E] Skåne University Hospital, Department on Hematology, Oncology and Radiation Physics, Lund, Sweden, [F] Department of Oncology, Copenhagen University Hospital - Rigshospitalet, Copenhagen, Denmark, [G] Department of Oncology, Experimental Cancer Therapy Unit Herlev Hospital, Herlev, Denmark, [H] University Hospitals of Leicester NHS Trust, Leicester, United Kingdom, [I] University Hospital Southampton NHS Foundation Trust, Southampton, United Kingdom, [J] BioInvent International AB, Lund, Sweden, [K] Merck & Co., Inc., Rahway, NJ, USA, [L] Centre for Cancer Immunology, University of Southampton, Southampton, United Kingdom

From Modest to Meaningful:

Targeting TNFR2 to Unlock Checkpoint Inhibitor Efficacy.

Enhancing PD-1 Blockade in Recurrent Ovarian Cancer.

Background

Advanced ovarian cancer still carries a poor prognosis: even with standard platinum-based chemotherapy, up to 80% of patients progress and treatment options are limited. Checkpoint inhibitor monotherapy has shown only modest benefit in recurrent disease (pembrolizumab; KEYNOTE-100 ORR 8%). However, pembrolizumab added to weekly paclitaxel ± bevacizumab (KEYNOTE-B96) improves PFS and OS in PD-L1 CPS ≥1 and all-comer patients (mPFS 8.3 vs 6.4 months/mOS 17.7 vs 14.0 months).

BI-1808 is an IgG1 mAb targeting TNFR2. It blocks TNF-α binding and, via FcγR engagement, depletes Tregs and reprograms myeloid cells, expanding antitumor CD8+ T cells. BI-1808 has shown promising monotherapy efficacy signals in both solid and hematological malignancies.

Preclinical models show that combining BI-1808 + aPD-1 in preclinical models leads to additive tumor inhibition, leading to exploration of BI-1808 combination with pembrolizumab in the clinical setting.

We present here interim results from an ongoing study in recurrent ovarian cancer with 17 patients on monotherapy and 25 patients on combination therapy. Data cut-off was April 20th 2026. An additional 9 patients on study have yet not reached first evaluation.

Study Overview

- Safety and efficacy of BI-1808 monotherapy or in combination with pembrolizumab are being evaluated in patients with advanced pretreated ovarian cancer.
- The study aims at enrolling 20 patients in monotherapy and 40 in combination. 12 patients under treatment in the combination cohort are still awaiting first assessment.
- All subjects enrolled were recurrent ovarian cancer patients, previously treated with at least one platinum containing treatment.
- Patients were treated with BI-1808 1000 mg +/- pembrolizumab 200 mg Q3W for up to two years.

Rationale

BI-1808 is an IgG1 mAb targeting TNFR2. It blocks TNF-α binding and, via FcγR engagement, depletes Tregs and reprograms myeloid cells, with a resulting expansion of antitumor CD8+ T cells. In preclinical models BI-1808 synergizes with anti-PD-1 leading to an additive tumor inhibition.

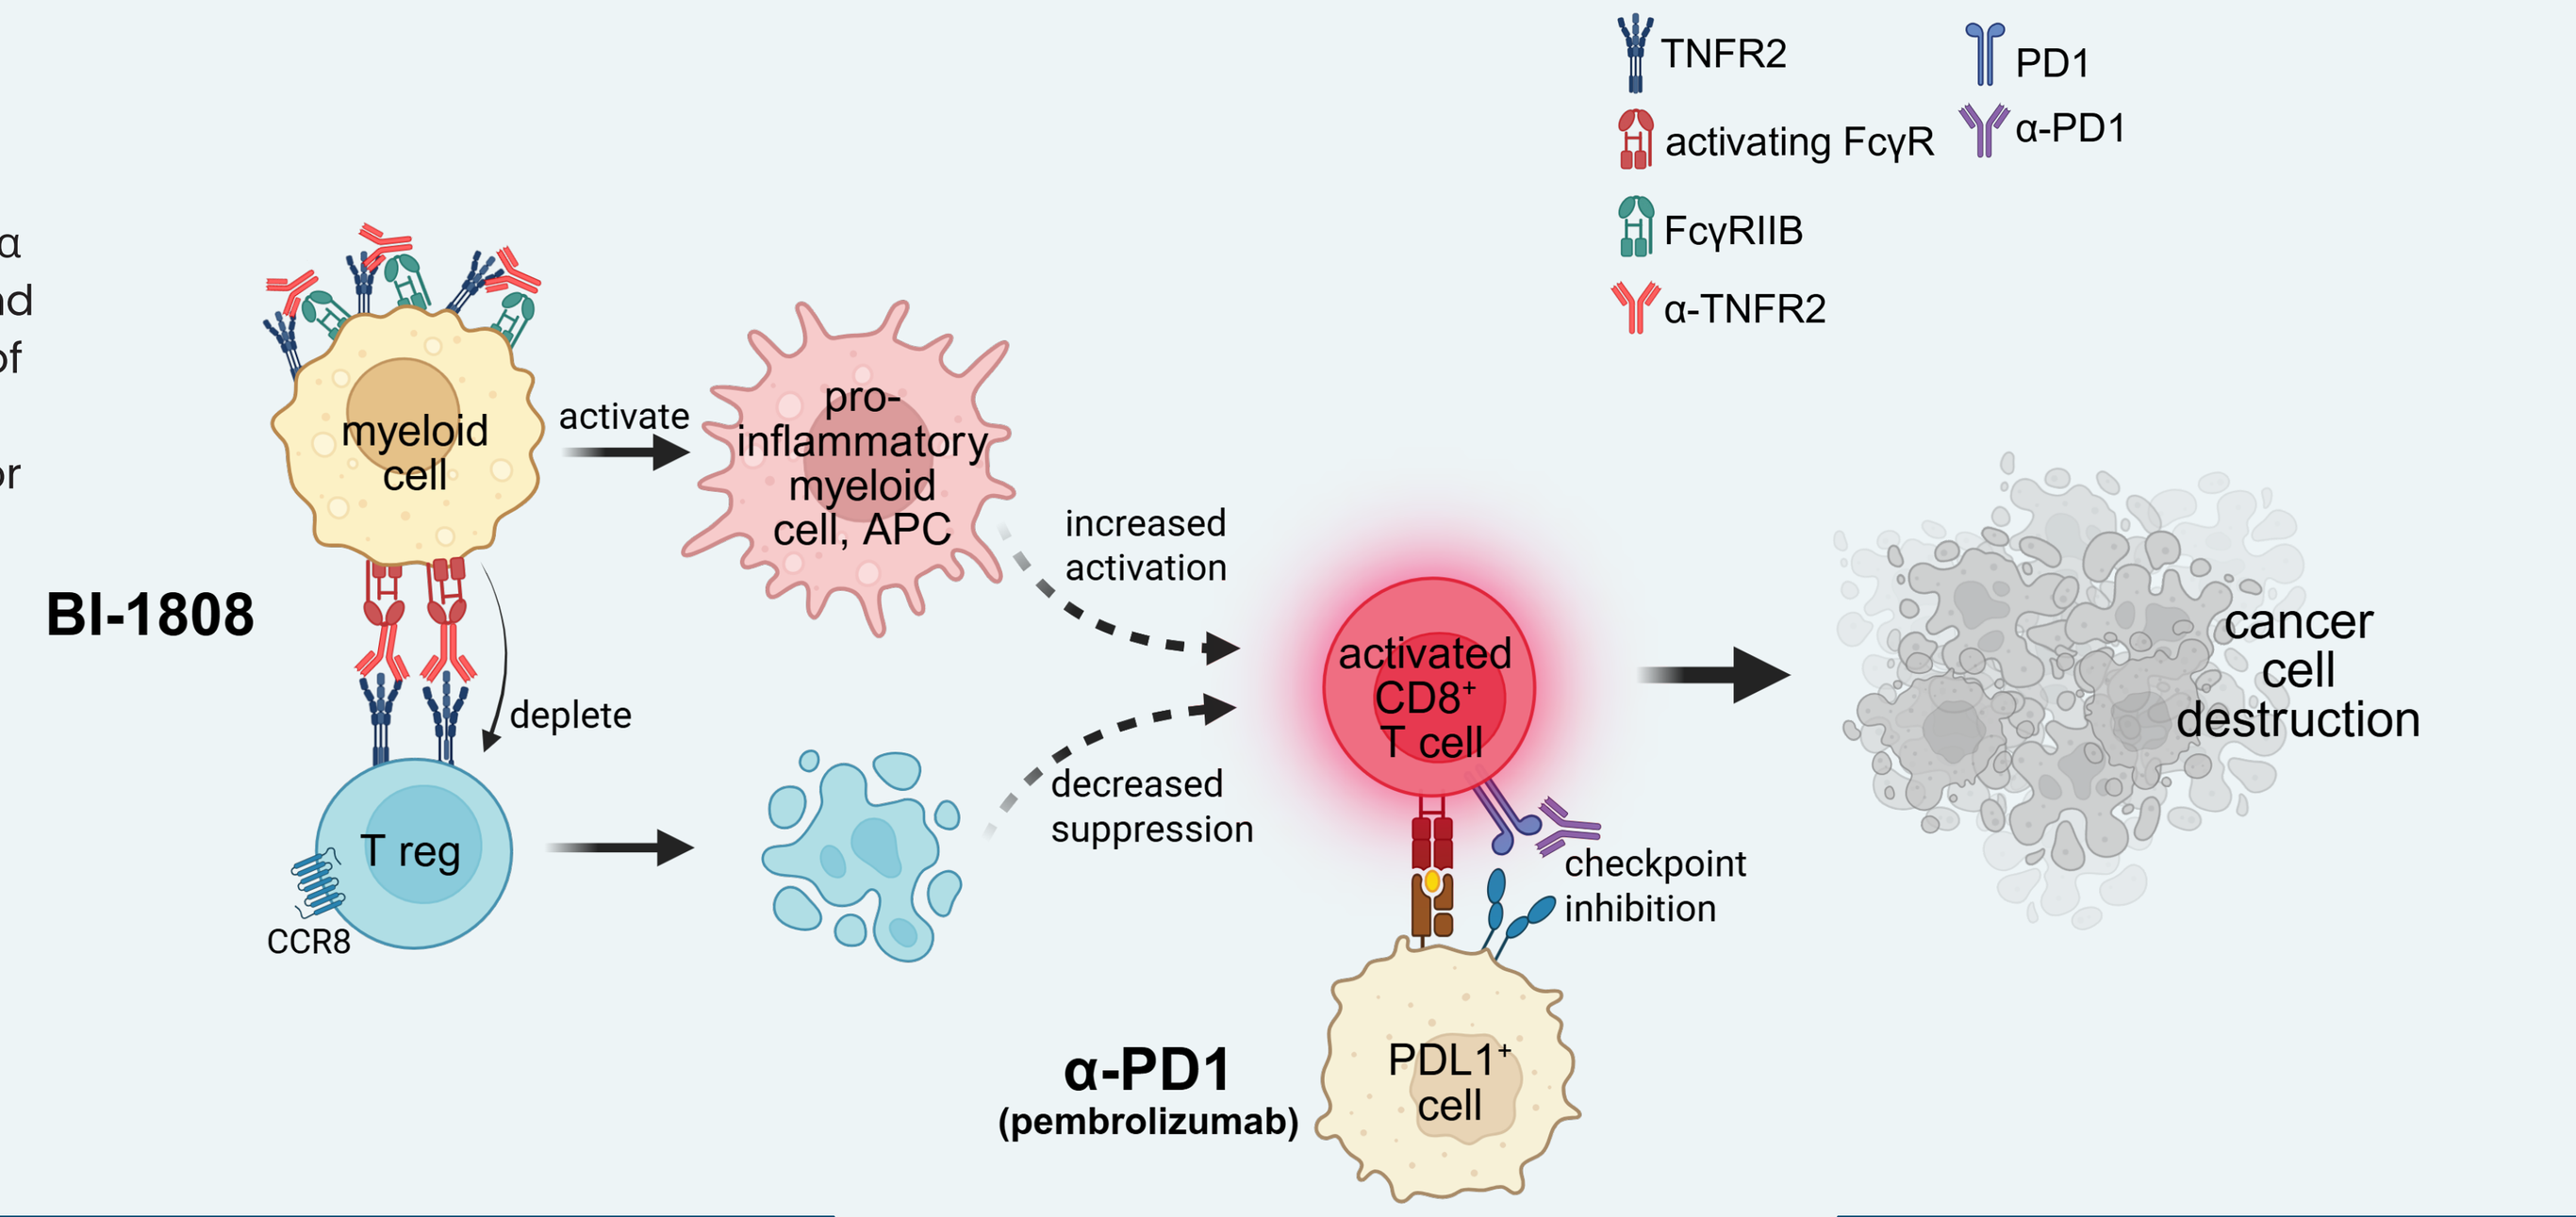
TNFR2
PD1
activating FcγR
α-PD1
FcγRIIB
α-TNFR2
BI-1808
myeloid cell
activate
pro-inflammatory myeloid cell, APC
increased activation
deplete
T reg
CCR8
decreased suppression
activated CD8+ T cell
checkpoint inhibition
cancer cell destruction
α-PD1 (pembrolizumab)
PDL1+ cell

Monotherapy

BI-1808 | Phase 2a Part A

Patient Population and Characteristics

The cohort comprised 17 subjects, including predominantly high-grade serous adenocarcinoma (13/17), with additional cases of clear-cell, serous papillary, endometriod, and carcinosarcoma subtypes.

Ovarian cancer subtype	Stage at initial diagnosis	Age (yrs.)	Prior treatment lines (no.)	Best response on BI-1808
ccOC	IIIA	63	4	iCR
HGSA	IV	79	8	iSD
Serous papillary carcinoma	IIIB	57	5	iSD
HGSA	UNK	75	6	iSD
HGSA	IV	66	4	iSD
HGSA	IV	57	5	iSD
HGSA	IIIB	54	7	iSD
HGSA	IV	59	5	iUPD
HGSA	IV	61	6	iUPD
HGSA	IV	67	3	iUPD
HGSA	IV	60	5	iUPD
HGSA	IV	42	5	iUPD
ccOC/endometrioid	IIIA	42	5	iUPD
Carcinosarcoma	IIIB	61	3	iUPD
HGSA	UNK	62	9	iUPD
HGSA	IV	39	4	iUPD
HGSA	IIIB	54	5	iUPD

Efficacy

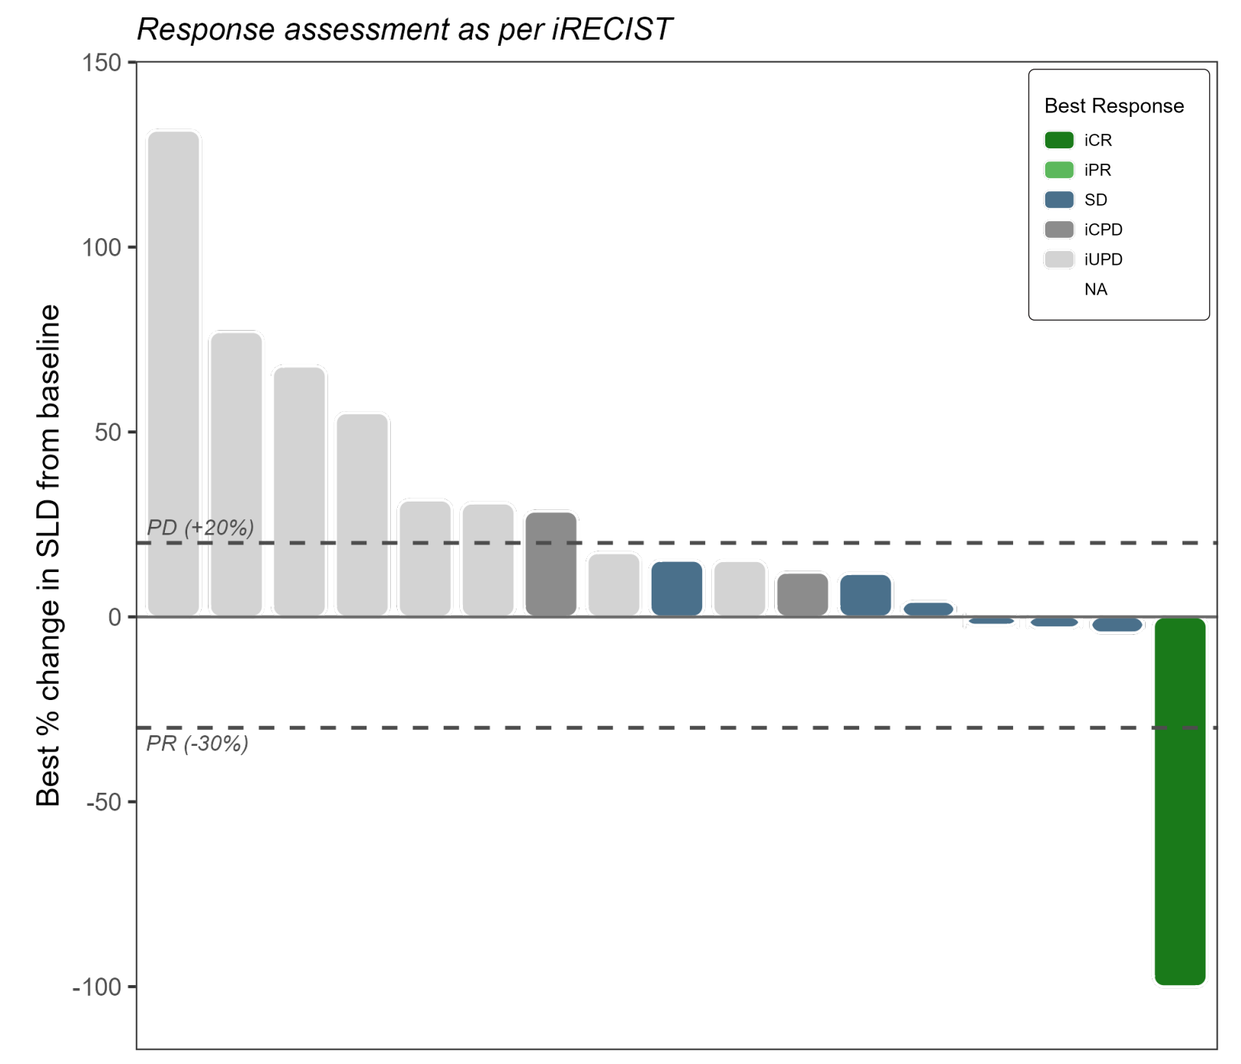
Response assessment as per iRECIST
Best Response
iCR
iPR
SD
iCPD
iUPD
NA
Best % change in SLD from baseline
PD (+20%)
PR (-30%)

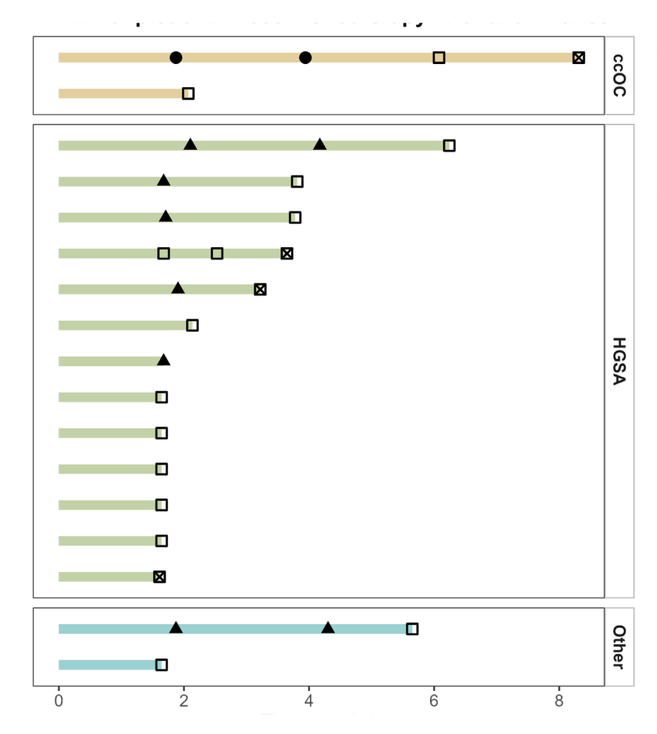
Indication
ccOC
HGSA
Other
Response
iCPD
iUPD
iCR
SD
NA

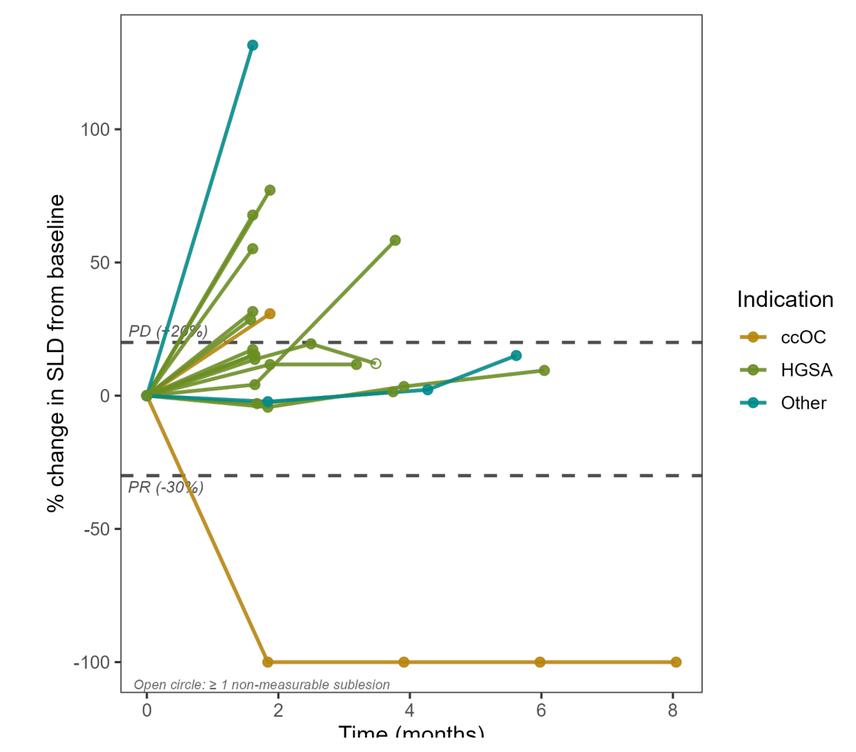
% change in SLD from baseline
PD (+20%)
PR (-30%)
Indication
ccOC
HGSA
Other
Open circle: ≥ 1 non-measurable sublesion
Time (months)

BI-1808 administration exhibits limited monotherapy activity

- 1 Complete Response observed 41% DCR (1 CR, 6 SD)
- 41% disease control rate demonstrated across subtypes

Safety Overview

BI-1808 treatment is well tolerated

- To date 62 patients with solid cancer have been administered BI-1808 as single agent
- No Grade 3+ events have been observed in monotherapy arm
- No treatment discontinuation due to safety events

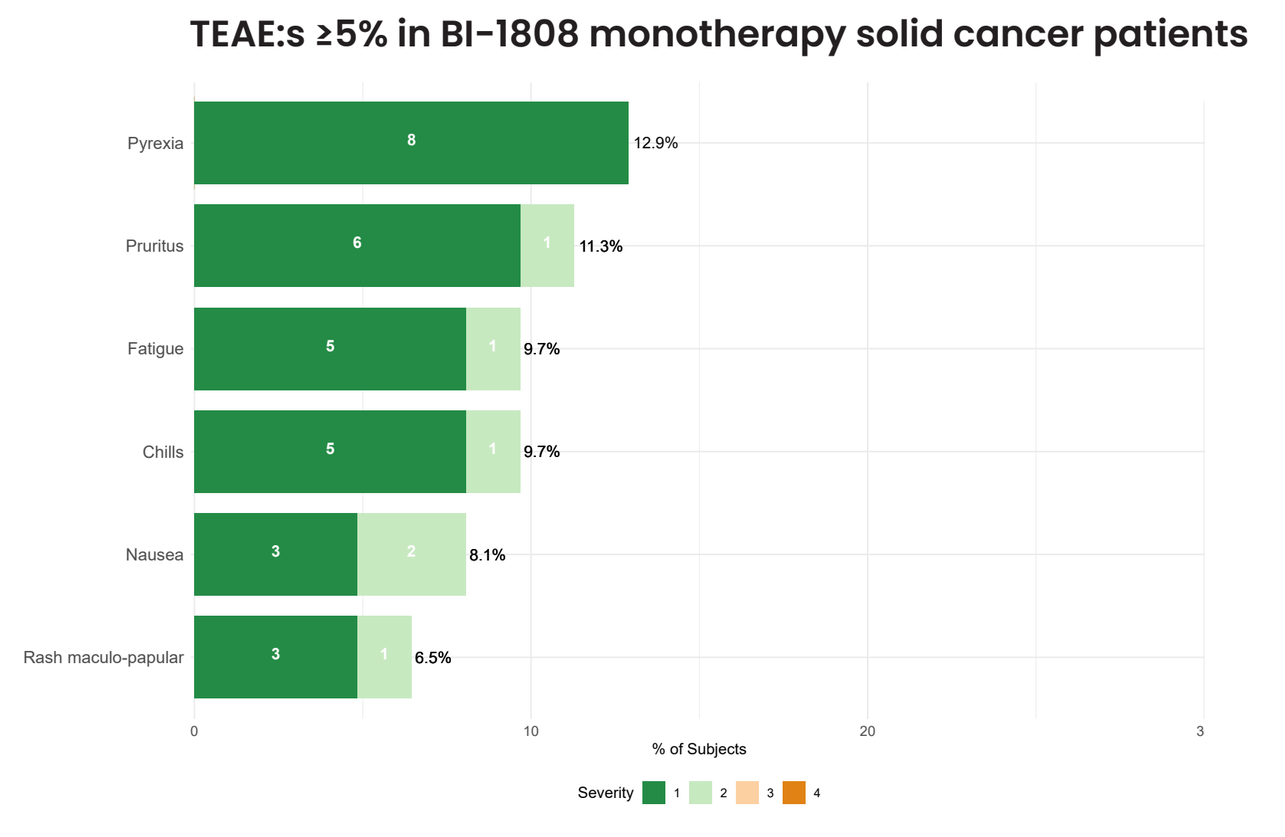
TEAE:s ≥5% in BI-1808 monotherapy solid cancer patients
Pyrexia 8 12.9%
Pruritus 6 1 11.3%
Fatigue 5 1 9.7%
Chills 5 1 9.7%
Nausea 3 2 8.1%
Rash maculo-papular 3 1 6.5%
% of Subjects
Severity 1 2 3 4

Combination Therapy

BI-1808 + pembrolizumab | Phase 2a Part B

Patient Population and Characteristics

The cohort comprised 26 subjects, including predominantly high-grade serous adenocarcinoma (17), with the remaining cases representing clear-cell ovarian carcinoma.

Ovarian cancer subtype	Stage at initial diagnosis	Age (yrs.)	Prior treatment lines (no.)	Best response on BI-1808+ pembrolizumab
ccOC	IIB	50	2	iCR
HGSA	IIIB	63	4	iPR
HGSA	IV	61	6	iPR
ccOC	UNK	61	3	iPR
ccOC	IV	67	4	iPR
ccOC	IB	79	4	iPR
HGSA	IV	61	7	iSD
HGSA	IV	64	7	iSD
HGSA	IIIB	75	5	iSD
HGSA	IIIB	75	9	iSD
ccOC	IIIB	57	1	iSD
ccOC	IA	46	1	iSD
HGSA	IV	62	5	iSD
HGSA	UNK	72	10	iCPD
HGSA	UNK	71	6	iUPD
HGSA	IV	65	4	iUPD
HGSA	IV	53	5	iUPD
ccOC	IB	62	2	iUPD
HGSA	IV	49	5	iUPD
HGSA	UNK	69	5	iUPD
HGSA	IIIA	54	7	iUPD
HGSA	IV	70	5	iUPD
ccOC	IIB	60	4	iUPD
ccOC	IV	59	3	iUPD
HGSA	IIIA	58	4	iUPD
HGSA	IV	68	9	NA

Efficacy

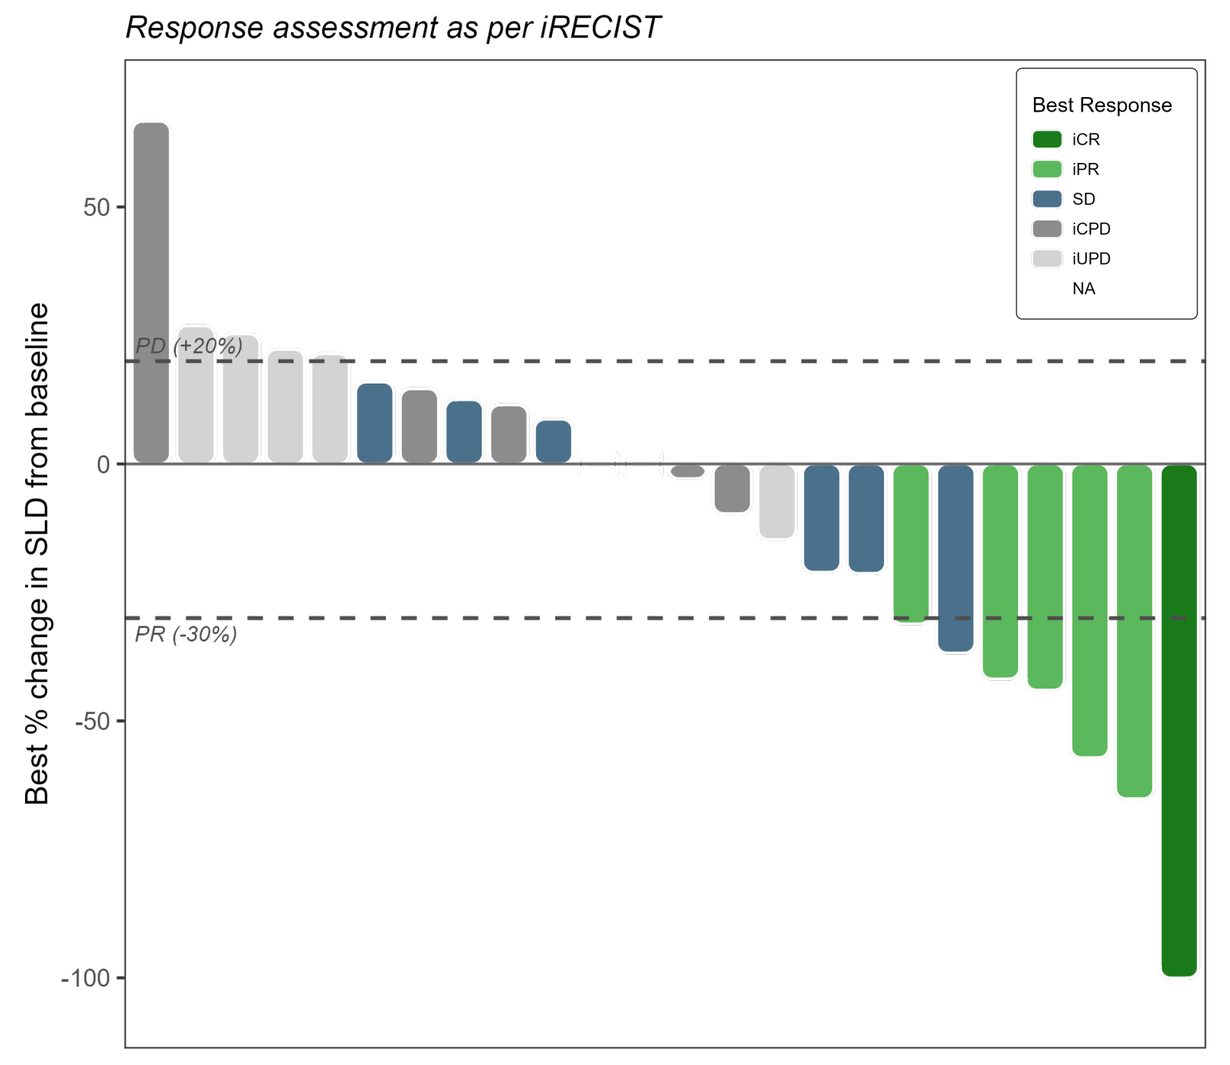
Response assessment as per iRECIST
Best Response
iCR
iPR
SD
iCPD
iUPD
NA
Best % change in SLD from baseline
PD (+20%)
PR (-30%)

One subject with iUPD lack reported post-baseline tumor size at data cut and not represented in waterfall plot

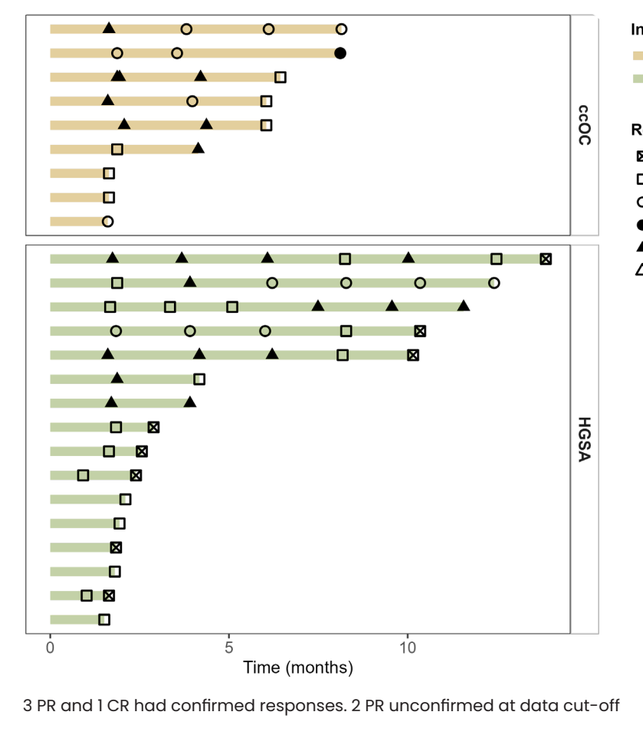
Indication
ccOC
HGSA
Response
iCPD
iUPD
iPR
iCR
SD
NA
Time (months)

3 PR and 1 CR had confirmed responses. 2 PR unconfirmed at data cut-off

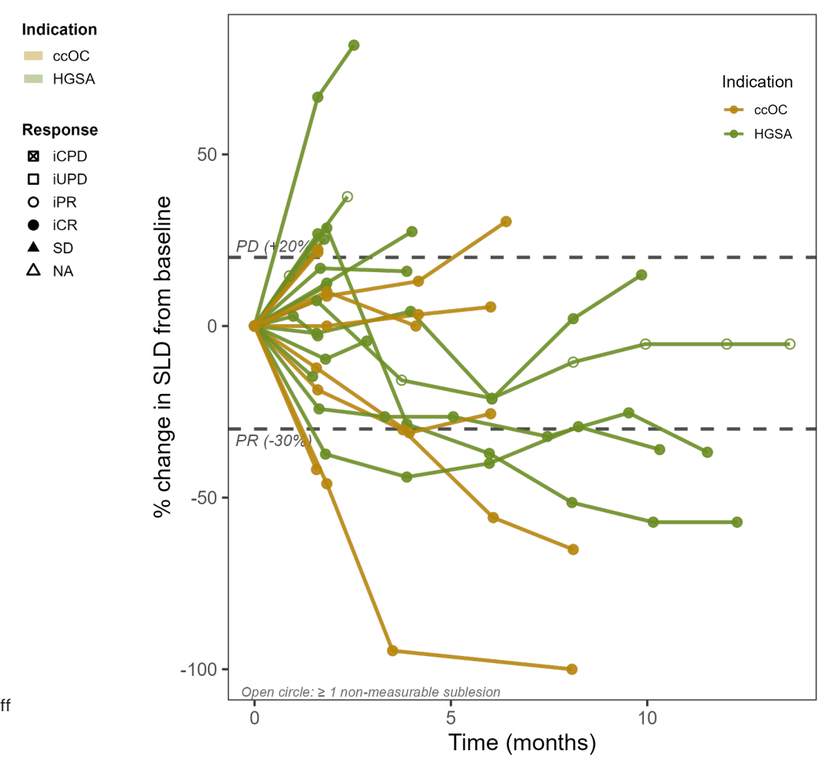
% change in SLD from baseline
Indication
ccOC
HGSA
PD (+20%)
PR (-30%)
Open circle: ≥ 1 non-measurable sublesion
Time (months)

We observe that lesions of some patients tend to increase in size initially before diminishing, possible evidence of pseudoprogression or delay in CD8+ T cell expansion. This finding is also seen in vivo preclinically.

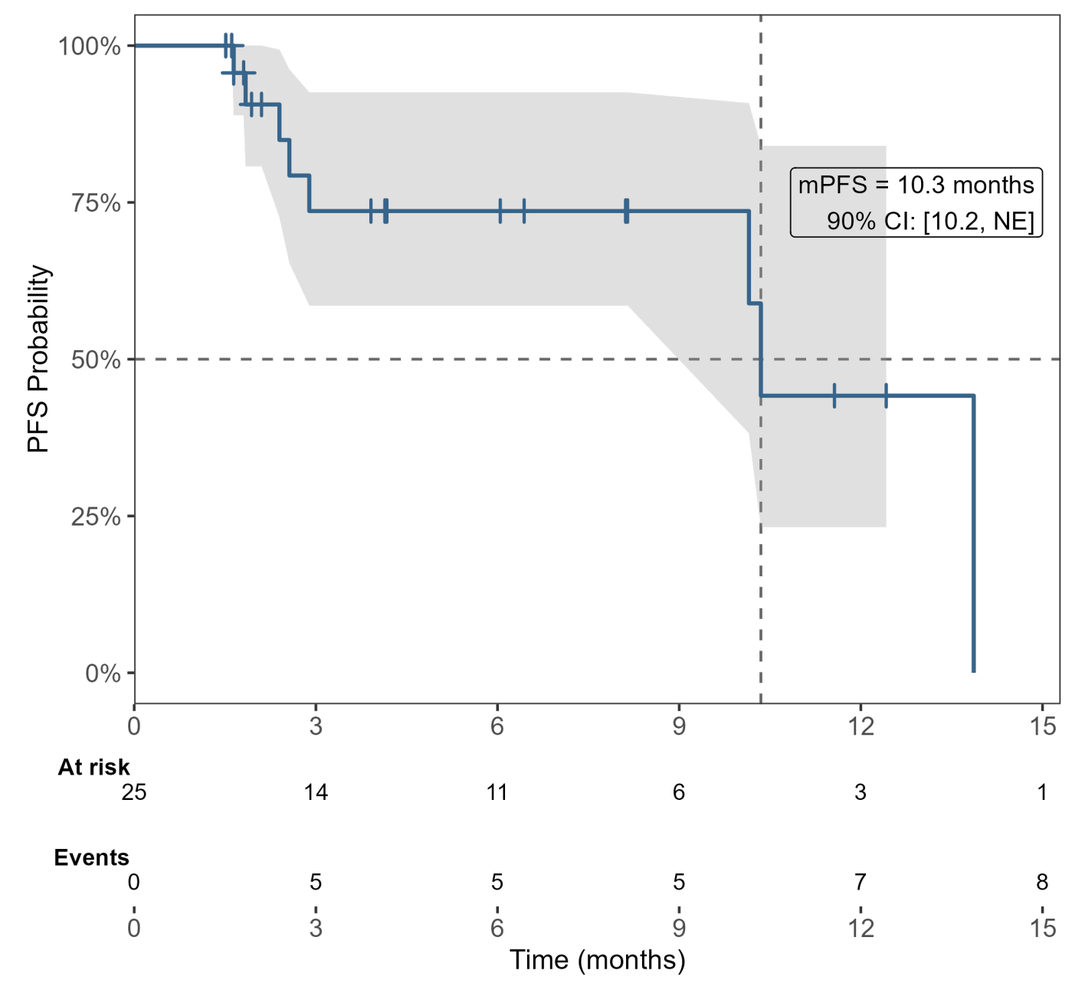
PFS Probability
mPFS = 10.3 months
90% CI: [10.2, NE]
At risk
25 14 11 6 3 1
Events
0 5 5 5 7 8
Time (months)

Kaplan-Meier plot and at-risk-tablefrom early PFS analysis in combinationcohort. 9 of these represented patients are still on treatment at cut-off, and mPFS will continue to mature.

- 24% ORR, 56% DCR
 - 1 CR
 - 5 PR
 - 8 patients with SD (several > 10 months)
- Responding patients in both high-grade serous and clear cell subtypes
- Preliminary analysis indicate a mPFS of 10.3 months

Safety Overview

Combination treatment is overall well tolerated

- To date 74 patients with solid cancer have been administered BI-1808+pembrolizumab
- Manageable immune related AE:s
- Low-rate of ≥ Grade 3+ AE:s
- Low-rate of treatment related discontinuations in combination arm (<5%)

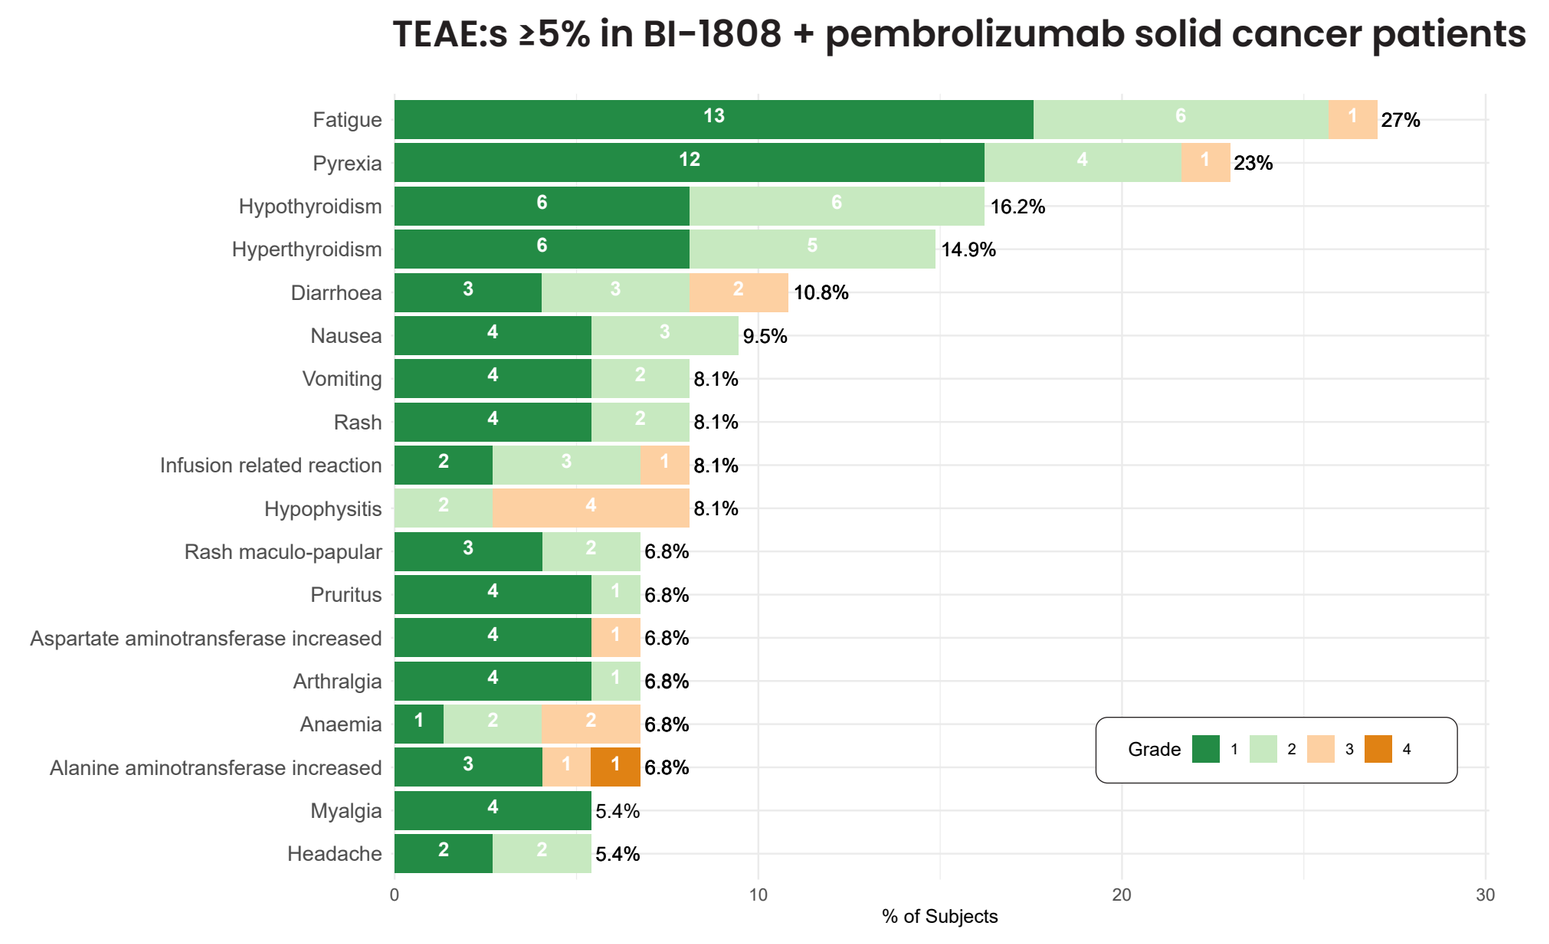
TEAE:s ≥5% in BI-1808 + pembrolizumab solid cancer patients
Fatigue 13 6 1 27%
Pyrexia 12 4 1 23%
Hypothyroidism 6 6 16.2%
Hyperthyroidism 6 5 14.9%
Diarrhoea 3 3 2 10.8%
Nausea 4 3 9.5%
Vomiting 4 2 8.1%
Rash 4 2 8.1%
Infusion related reaction 2 3 1 8.1%
Hypophysitis 2 4 8.1%
Rash maculo-papular 3 2 6.8%
Pruritus 4 1 6.8%
Aspartate aminotransferase increased 4 1 6.8%
Arthralgia 4 1 6.8%
Anaemia 1 2 2 6.8%
Alanine aminotransferase increased 3 1 1 6.8%
Myalgia 4 5.4%
Headache 2 2 5.4%
% of Subjects
Grade 1 2 3 4

Conclusions

- Pembrolizumab monotherapy in KEYNOTE-100 showed ORR of 8% and mPFS of 2.1 months in OC, and BI-1808 monotherapy exhibit similar limited activity with 1 CR and mPFS of 3.6 months.
- Data across preclinical and translational studies support evidence of suppressing protective mechanisms in ovarian cancer when combining BI-1808 with pembrolizumab.
- An objective response rate of 24% in the combination arm indicate a meaningful treatment benefit of BI-1808 + pembrolzumab combination in OC patients.
- 56% of patients exhibit disease control, and an early PFS measure of mPFS=10.3 months consitutes a very promising efficacy signal, paired with favorable safety profile.
- Translational data show BI-1808 mediated deletion of regulatory T cells and myeloid reprogramming as measured by induction of IL-12. When combined with pembrolizumab, CD8+ T cells are activated and cytokines indicative of TLS formation are increased.
- Preclinical data indicate that time to response is longer than typically observed with immunecheckpoint therapy. This suggests that additional delayed responders may have been missed at initial w9 scan. ctDNA will be closely followed in upcoming patients.

Translational Data

BI-1808 induces T reg depletion and CD8+ T cell activation

Regulatory T cells strongly suppress CD8+ T cells, which are crucial for anti-tumor immune responses. BI-1808 reduces regulatory T cells and, when combined with pembrolizumab, increase levels of activated CD8+ T cells.

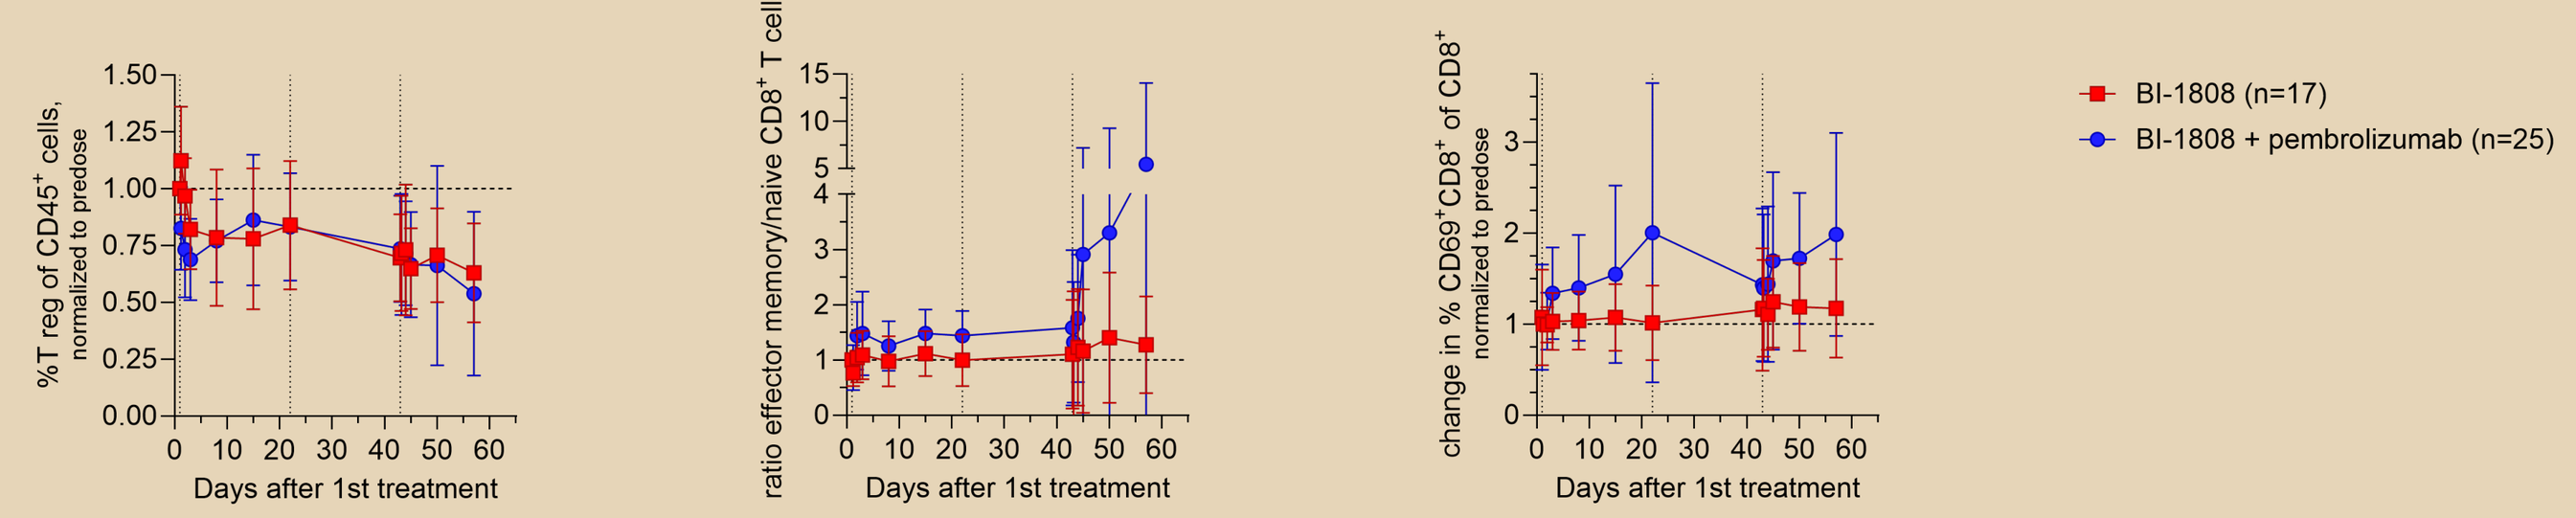
% T reg of CD45+ cells, normalized to predose
ratio effector memory/naive CD8+ T cells
change in % CD69+CD8+ of CD8+ normalized to predose
Days after 1st treatment
BI-1808 (n=17)
BI-1808 + pembrolizumab (n=25)

T reg levels (measured as % T regs of total CD45+ cells) in blood of BI-1808 or BI-1808 + pembrolizumab treated patients during the first 3 treatment cycles. Dotted line indicate pre dose baseline mean value.

Ratio of effector memory to naive CD8+ T cells and change in % CD69+CD8+ T cells of total CD8+ T cells in blod of BI-1808 (N=17) or BI-1808 + pembrolizumab treated (N=25) patients during the first 3 treatment cycles. Dotted line indicate pre dose baseline mean value.

BI-1808 induces cytokines associated with CD8+ T cell activation, myeloid proinflammatory reprogramming and formation of tertiary lymphoid structures

IFN-gamma and CXCL9 are markers of CD8+ T cell activation shown to be essential for antitumor immunity.

Interleukin-12 (IL-12) is a potent immunomodulator in the tumor microenvironment (TME), reversing the immunosuppressive activity of tumor-associated macrophages (TAMs), reducing the M2 phenotype while promoting the M1 phenotype and allowing more efficacious presentation of tumor antigens.

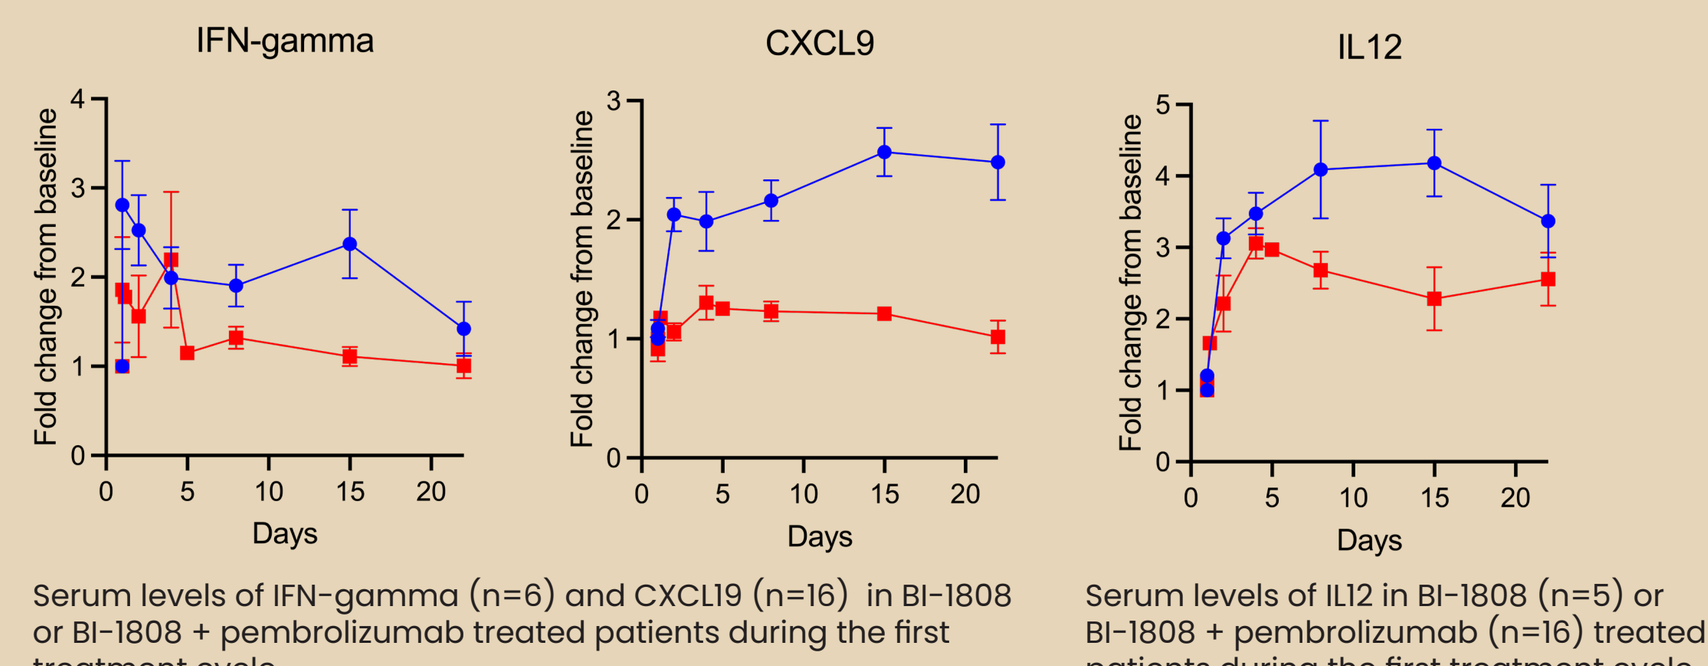
IFN-gamma
CXCL9
IL12
Fold change from baseline
Days

Serum levels of IFN-gamma (n=6) and CXCL19 (n=16) in BI-1808 or BI-1808 + pembrolizumab treated patients during the first treatment cycle.

Serum levels of IL12 in BI-1808 (n=5) or BI-1808 + pembrolizumab (n=16) treated patients during the first treatment cycle.

CCL19 and CXCL13 are key chemokines involved in the formation, structure, and function of Tertiary Lymphoid Structures (TLS) within the tumor microenvironment. TLS's are crucial for supporting and maintaining active anti-tumor T cell responses.

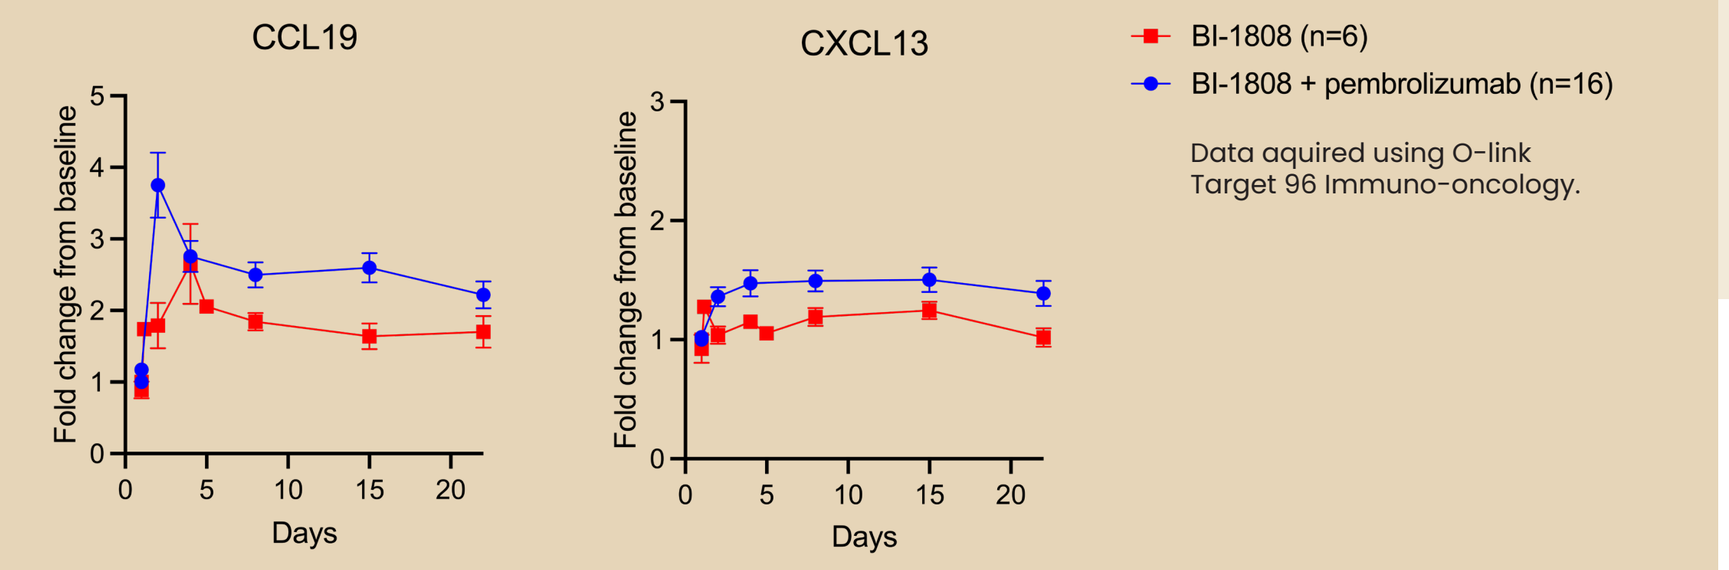
CCL19
CXCL13
Fold change from baseline
Days
BI-1808 (n=6)
BI-1808 + pembrolizumab (n=16)

Data aquired using O-link Target 96 Immuno-oncology.

Serum levels of CCL19 and CXCL13 in BI-1808 (n=6) or BI-1808 + pembrolizumab (n=16) treated patients during the first reatment cycle.

BI-1808 and Time to Response

- Prior pembrolizumab data in OC indicate a median time to response of 9-12 weeks
- Intrapolation of preclinical data give a predicted time to response of 19-25 weeks for BI-1808, and 13-17 weeks for BI-1808 + pembrolizumab
- 86% of PD exclusions occurred within 13 weeks, which may be too early to detect responses and could mistakenly interpret pseudoprogression as tumor progression

Preclinical time to response

- Compared to α-PD-1, response occurs later with mBI-1808
- Higher tumor growth before regression, but increase in survival
- mBI-1808 + α-PD-1 combination improve these parameters

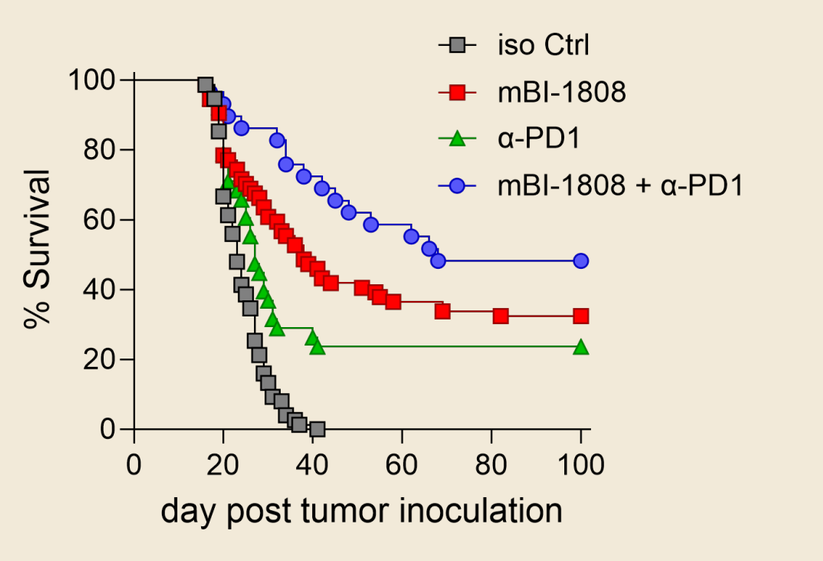
iso Ctrl
mBI-1808
α-PD1
mBI-1808 + α-PD1
% Survival
day post tumor inoculation

Survival in mice inoculated with EMT6 cancer cells (n=29-75/group).

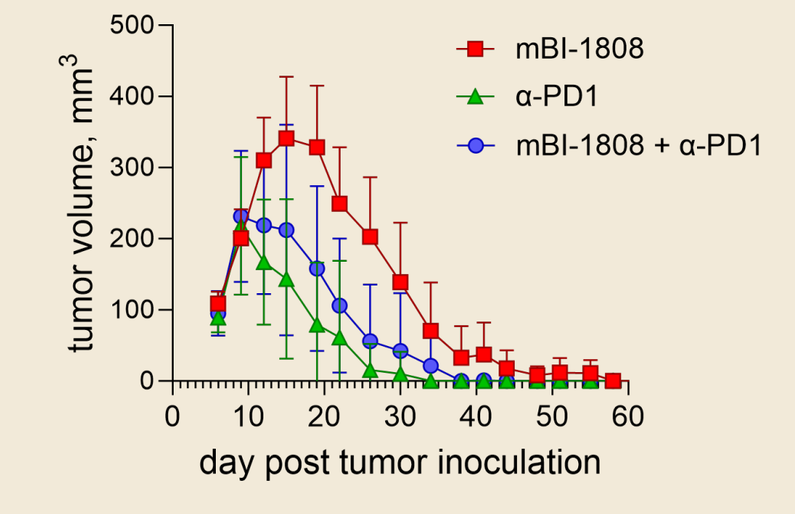
mBI-1808
α-PD1
mBI-1808 + α-PD1
tumor volume, mm3
day post tumor inoculation

Tumor dynamics of cured mice by mBI-1808 (n=20) or α-PD-1 (n=9) or mBI-1808 + α-PD-1 (n=13).

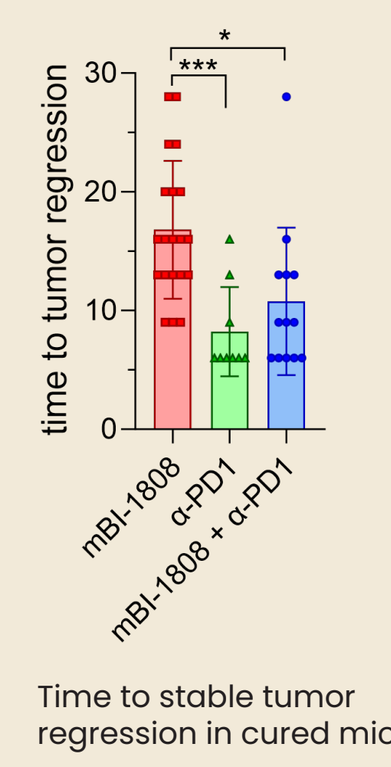
time to tumor regression

*
mBI-1808
α-PD1
mBI-1808 + α-PD1

Time to stable tumor regression in cured mice.

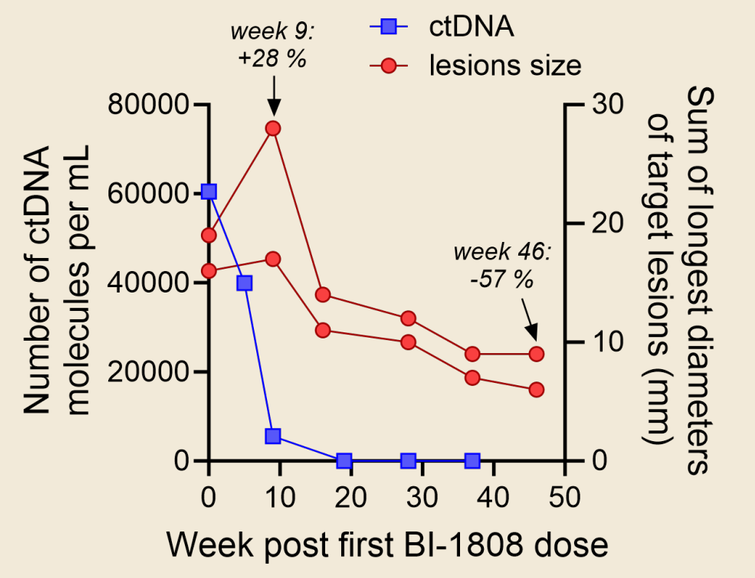
week 9: +28 %
week 46: -57 %
ctDNA
lesions size
Number of ctDNA molecules per mL
Sum of longest diameters of target lesions (mm)
Week post first BI-1808 dose

Clinical case: OC patient treated with BI-1808 + pembrolizumab

- First tumor scan at w9 show increase in both target lesion.
- Patient was kept on study, and at w16 both target lesions had decreased.
- ctDNA rapidly forecast response already at w5
- No ctDNA was detected at w27, and patient still on treatment at 1 year.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Contact information

Contact information: clincialtrials@bioinvent.com
Study identifier: NCT04752826

This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA

Acknowledgements

We extend our deep appreciation to study participants, their families and clinical research staff.